TRENDS IN CONTEMPORARY CARDIOVASCULAR RISK ASSESSMENT IN CANADA: RESULTS OF THE PARADIGM PERCEPTIONS STUDY

M. Gupta, [1,2,3] M. Tsigoulis, [1] M. Kajil, [1] S. Hirjikaka, [1] A. Quan, [3] H.Teoh [1,3] and S. Verma [1,3]

[1] Canadian Cardiovascular Research Network, Brampton, ON; [2] Division of Cardiology, McMaster University, Hamilton, ON; [3] Division of Cardiac Surgery, St Michael's Hospital, Toronto, ON

ABSTRACT

Objective: There are limited data in the contemporary era as to how primary care physicians (PCP) assess global cardiovascular risk and incorporate non-traditional risk factors in their decision making. **Methods:** We conducted a prospective cross-sectional survey of 652 Canadian PCP. Using a structured and validated tool, we evaluated their perceptions regarding risk assessment, lipid treatment thresholds, and novel biomarkers. **Results:** PCP indicated that they used the Framingham Risk Score (FRS) to estimate global risk in 67% of cases, yet 31% incorrectly identified a high-risk patient according to the FRS. Non-classical risk factors such as family history or central obesity were identified by 40% as the single most important determinant of initiating formal risk assessment. PCP reported that they reclassified patients into high-risk categories in the presence of metabolic syndrome or positive family history in 62% and 64% of cases respectively. Waist circumference (WC) was considered as a vital sign by 84% yet only 6% reported routinely documenting this. Inflammation was identified as the main mechanism of atherosclerosis by 50% of PCP, and 100% were aware of hsCRP as an additional risk marker. However, 51% were unclear about the clinical utility of this biomarker to risk stratify an intermediate risk patient beyond traditional risk factors, and 73% were unsure if elevated hsCRP identified a patient population requiring statin treatment. CT angiography was identified by 24% as the best test for screening in primary prevention. **Conclusions:** PCP incorporate non-traditional measures, in addition to FRS, in risk stratification. Although central obesity was identified as an important factor prompting formal risk assessment, WC was documented by fewer than 6% of PCP. The majority of respondents were unclear about the clinical utility of hsCRP in risk stratification or treatment. One quarter of PCP suggested that CTA was the best suited test for screening large populations.

INTRODUCTION

Appropriate cardiovascular risk stratification is imperative for the optimal application of preventive therapies. Despite the availability of evidence-based guidelines for cardiovascular prevention, studies continually demonstrate a persisting care gap in clinical medicine.

OBJECTIVE

To evaluate the current perceptions of primary care physicians regarding cardiovascular risk assessment and lipid treatment thresholds.

STUDY DESIGN

Participants: 2225 primary care physicians, from 10 Canadian provinces, were invited by mail between October 2008 and May 2009 to complete a validated 32-question survey aimed at determining their current practice of global cardiovascular risk assessment and the subsequent treatment algorithms employed.

Data Handling: Completed surveys were returned to the Canadian Cardiovascular Research Network where the entries were read with DataFax 3.9 software. The data were analyzed by two investigators (H.T. and A.Q.) who were blinded to the identities of the physicians.

RESULTS

The blue bar in each figure denotes the most correct answer (where applicable).

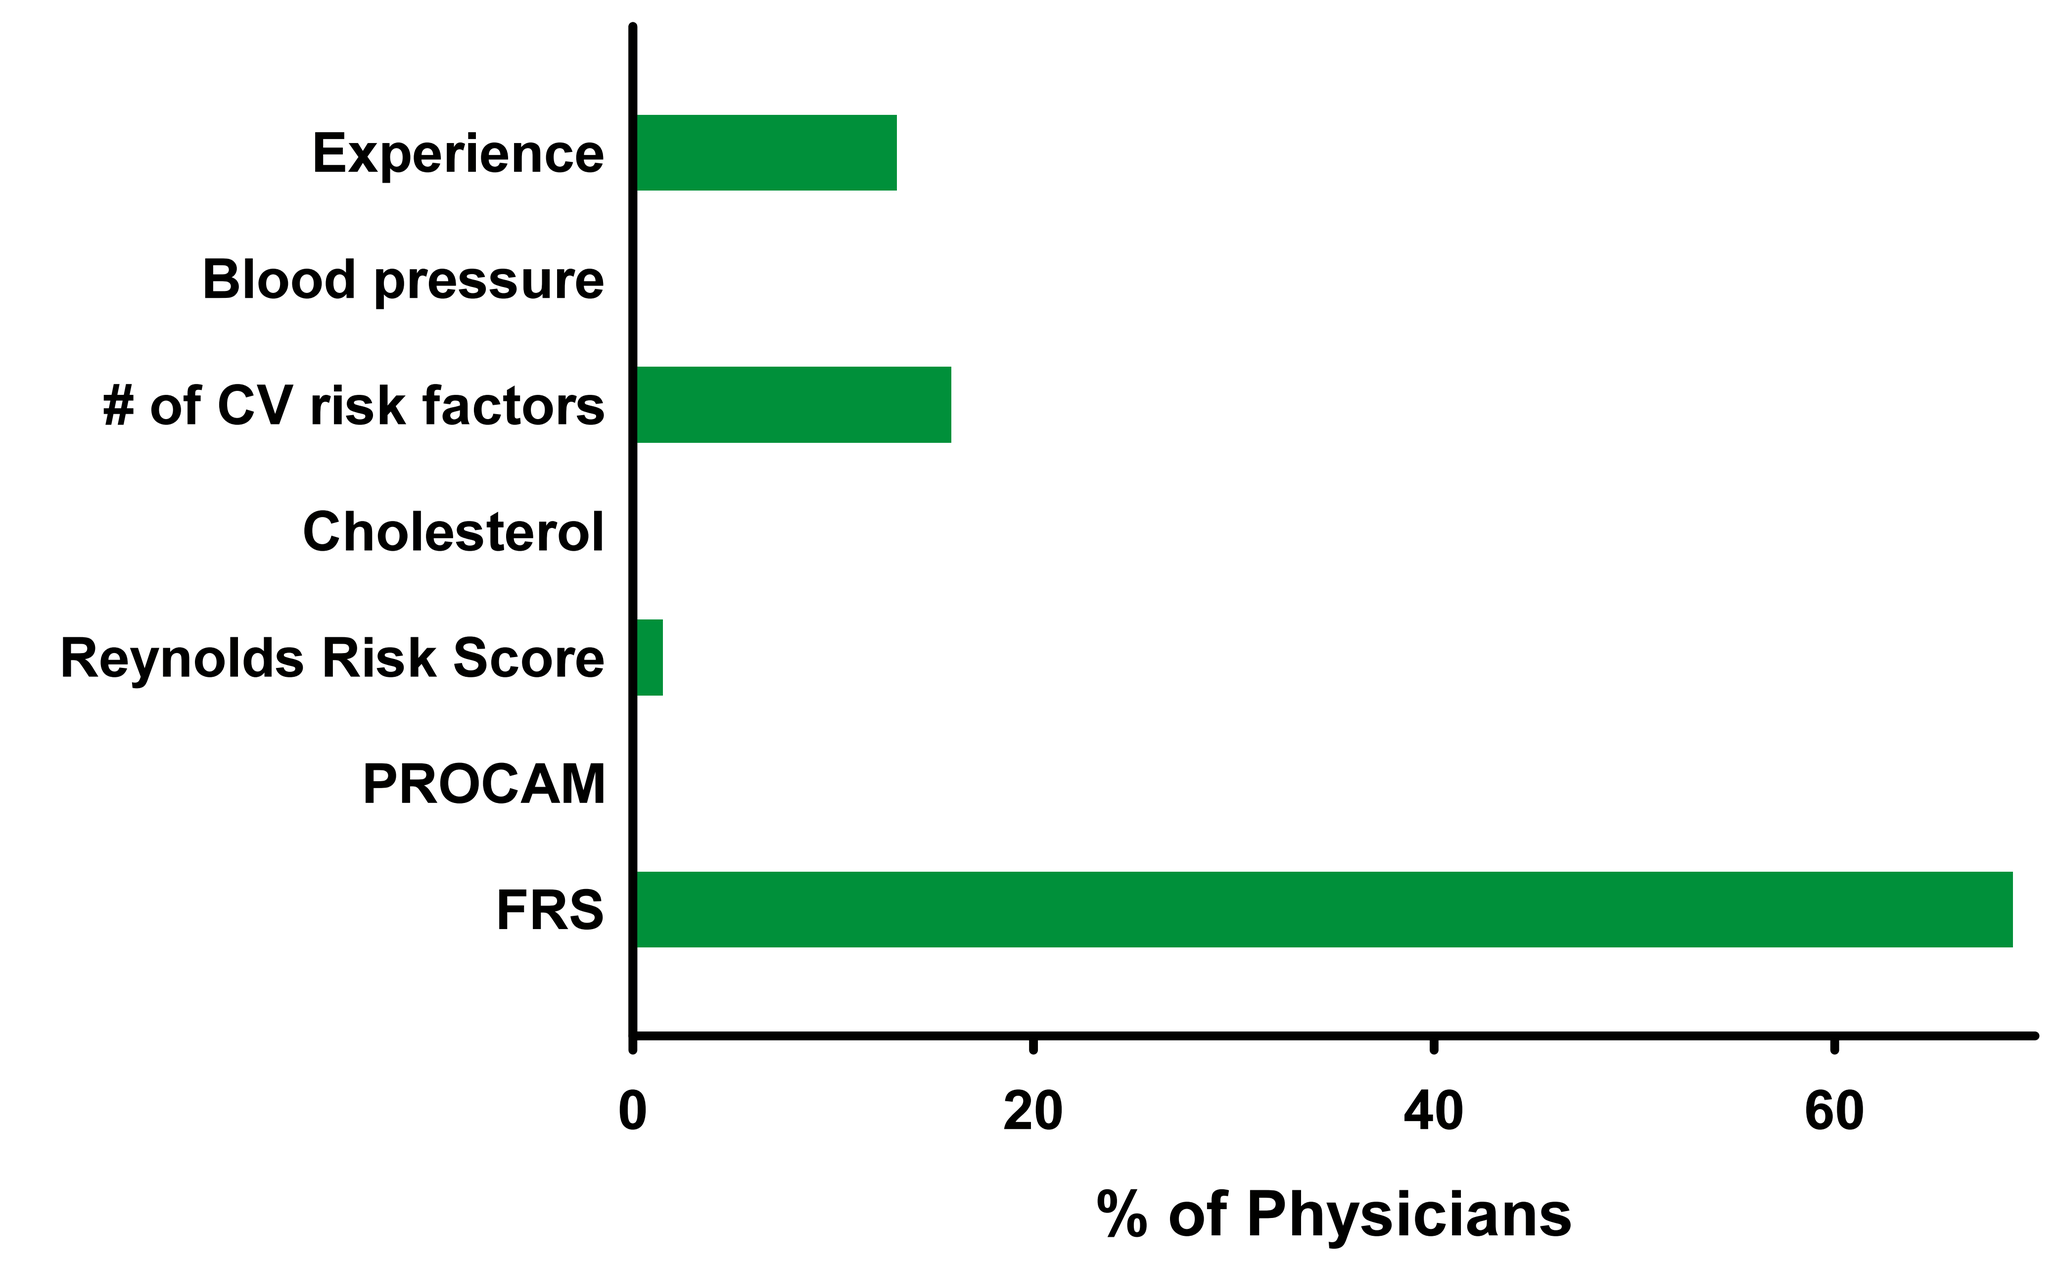
Experience
Blood pressure
of CV risk factors
Cholesterol
Reynolds Risk Score
PROCAM
FRS
0
20
40
60
% of Physicians

Fig. 1. Responses to "How do you estimate cardiovascular risk in clinical practice?"

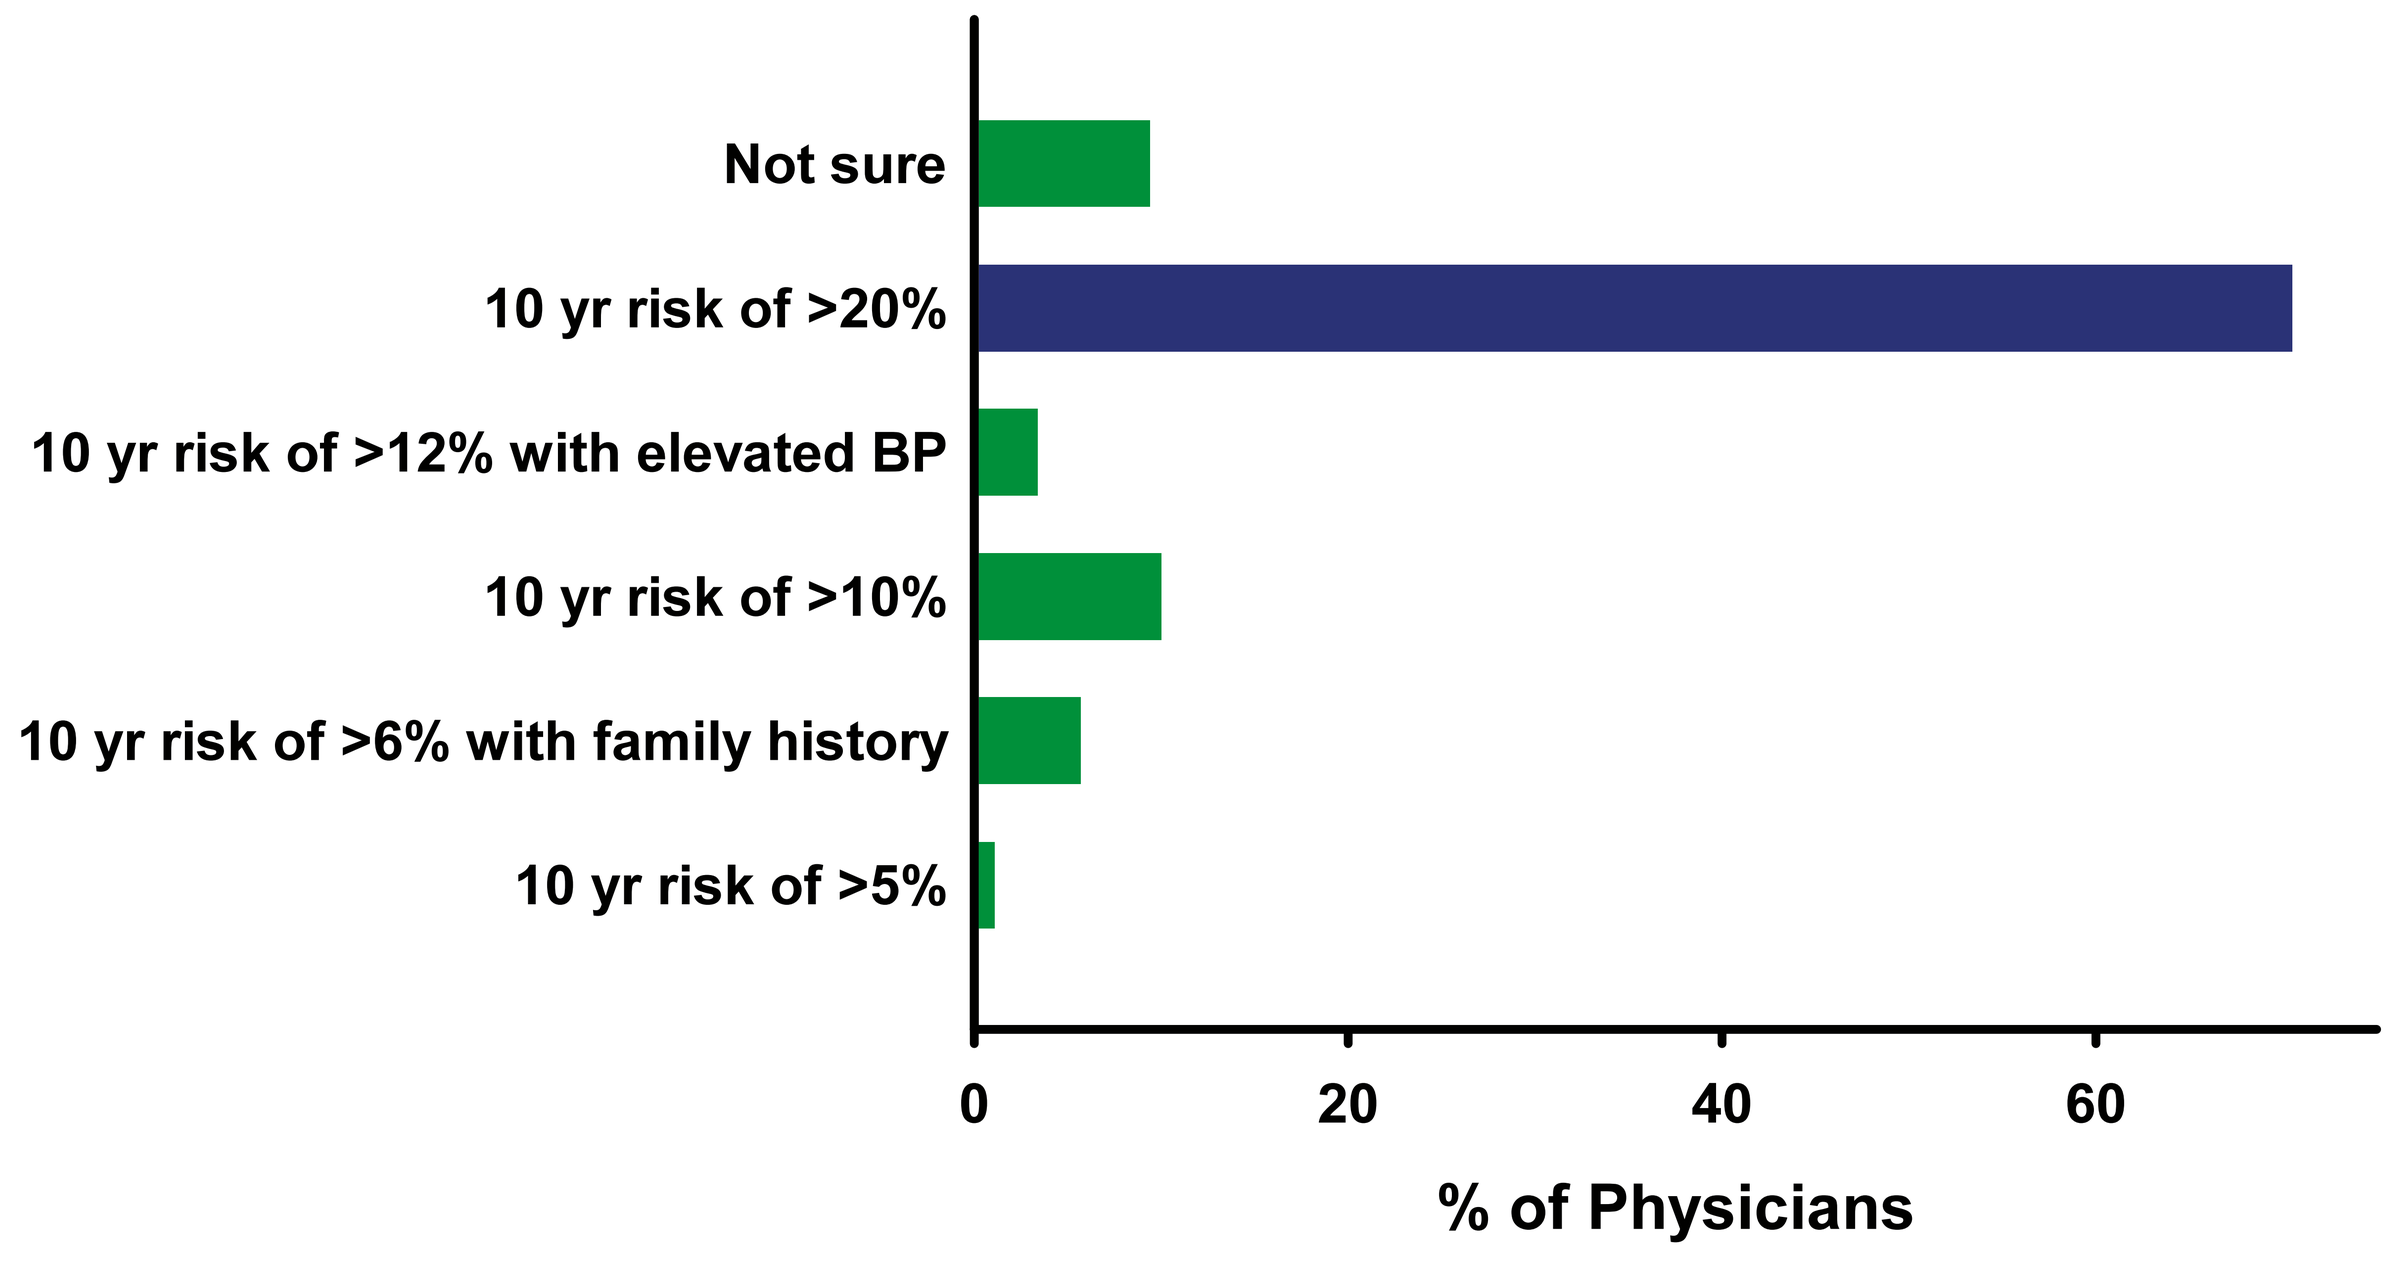
Not sure
10 yr risk of >20%
10 yr risk of >12% with elevated BP
10 yr risk of >10%
10 yr risk of >6% with family history
10 yr risk of >5%
0
20
40
60
% of Physicians

Fig. 2. Responses to "Which one of the following options is consistent with high risk as estimated by the FRS?"

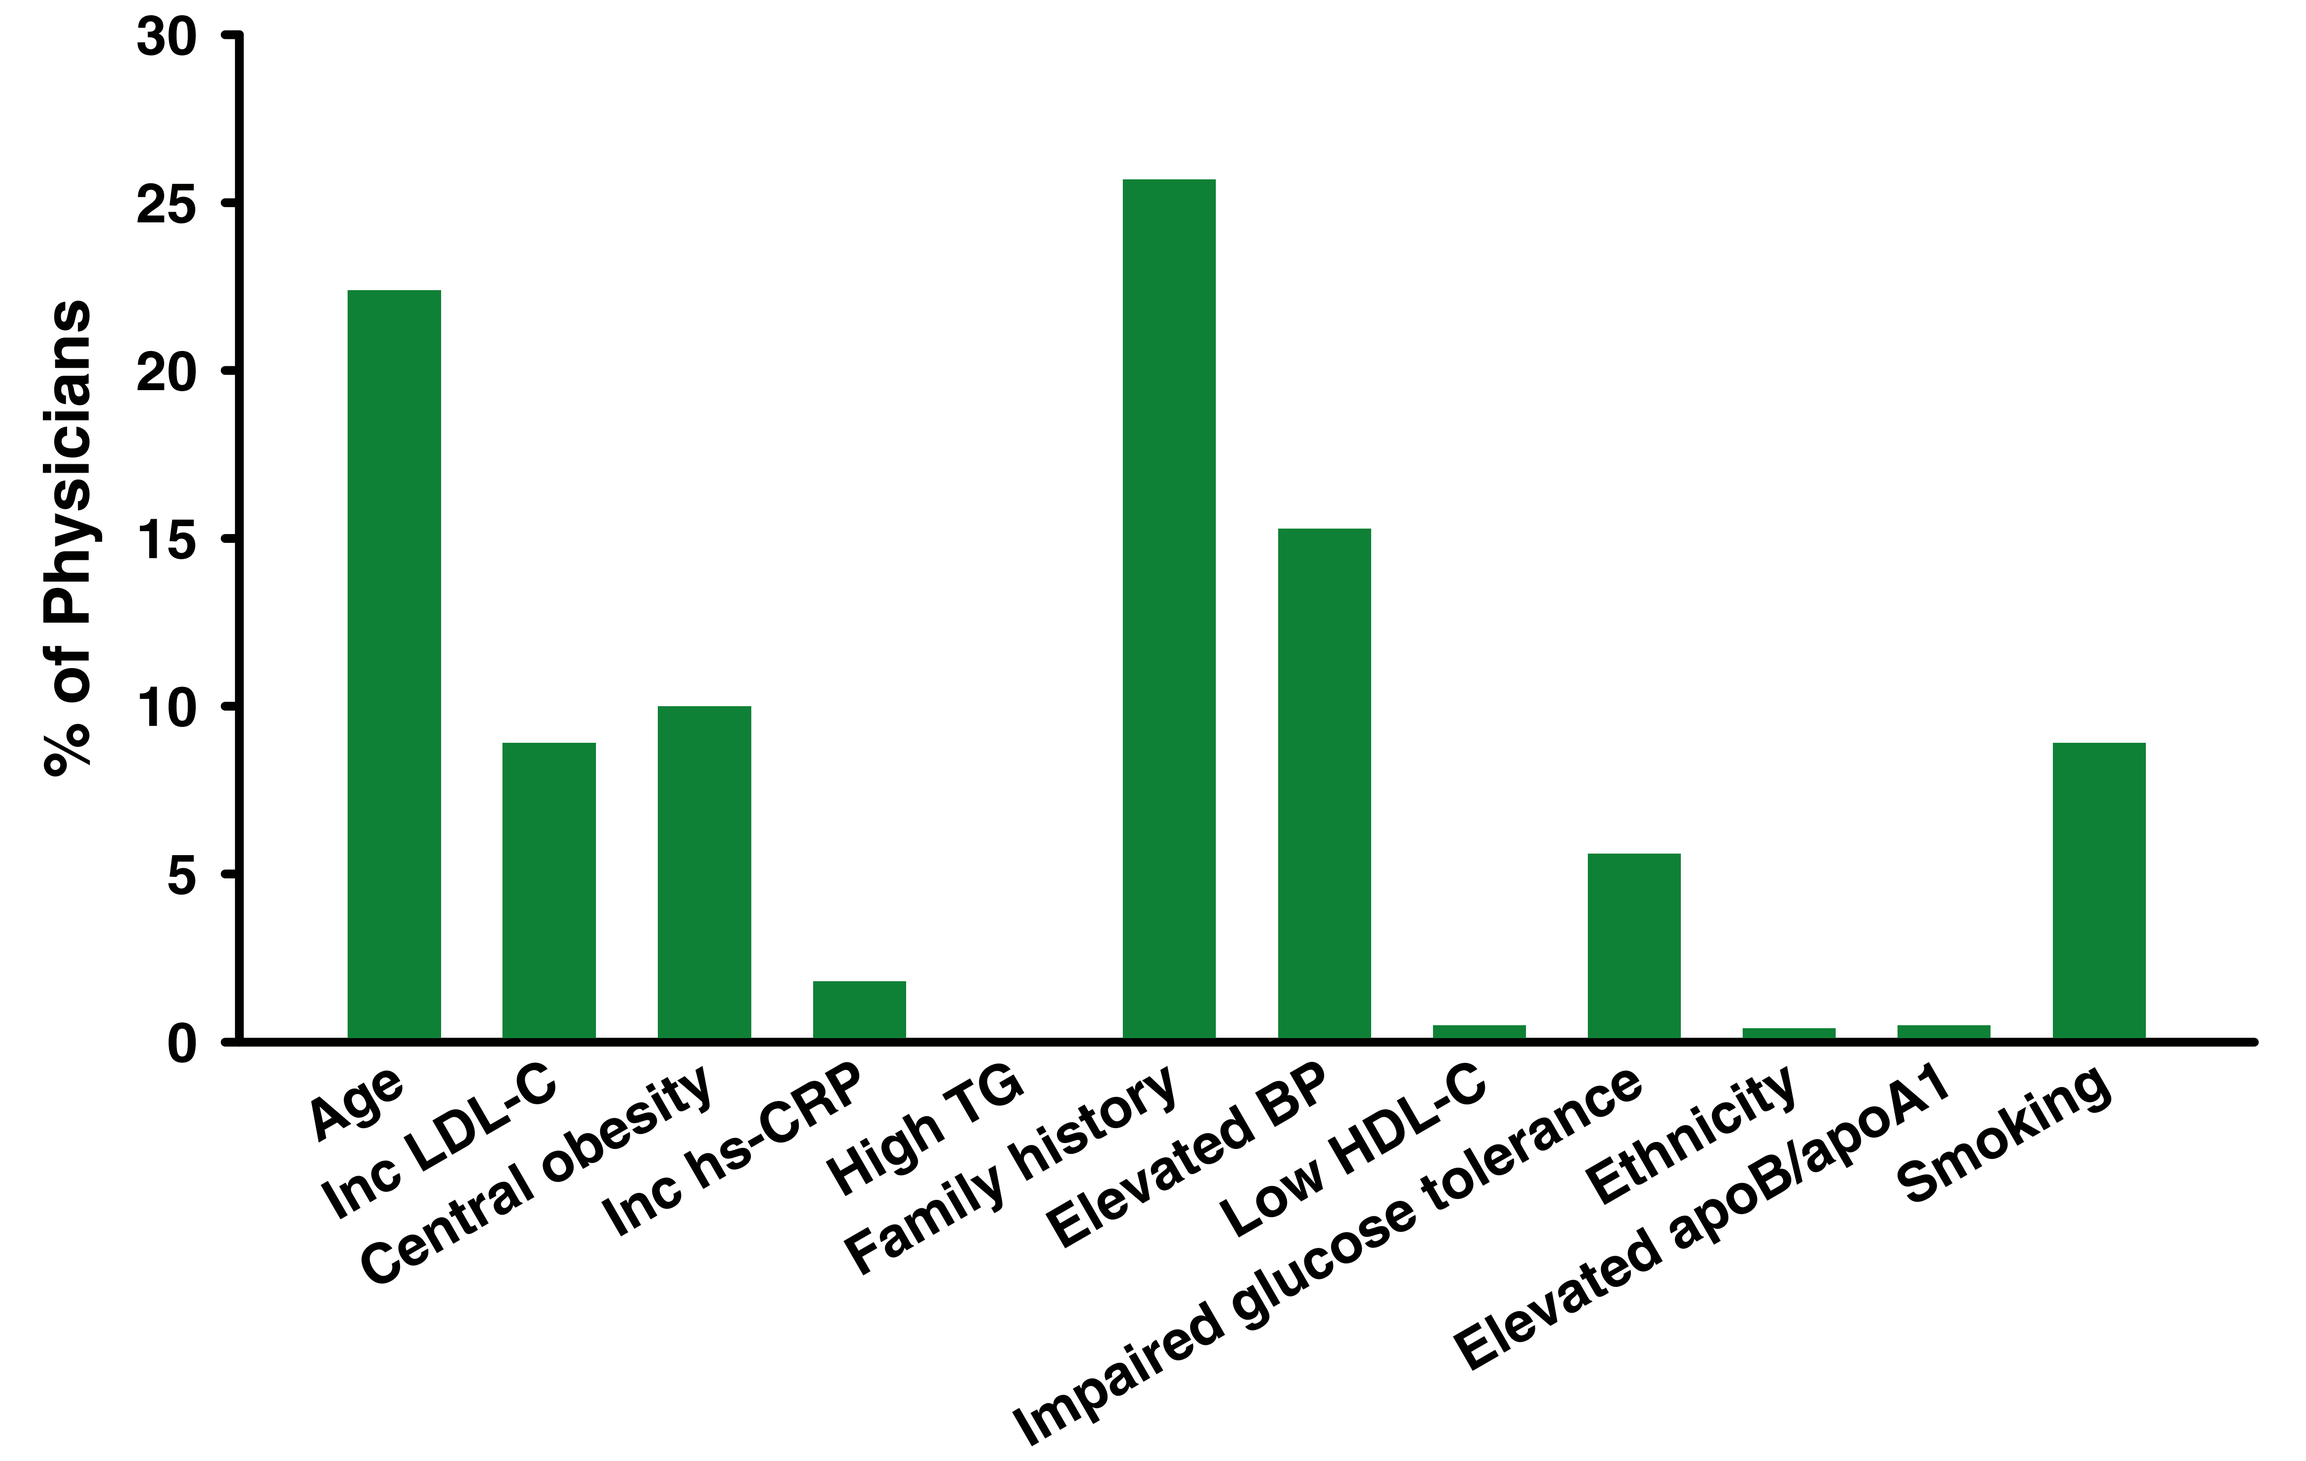
% of Physicians
0
5
10
15
20
25
30
Age
Inc LDL-C
Central obesity
Inc hs-CRP
High TG
Family history
Elevated BP
Low HDL-C
Impaired glucose tolerance
Ethnicity
Elevated apoB/apoA1
Smoking

Fig. 3. Responses to "In an asymptomatic patient, please mark the <u>single</u> factor you consider to be the <u>most</u> important in making your decision to perform a formal cardiovascular risk assessment."

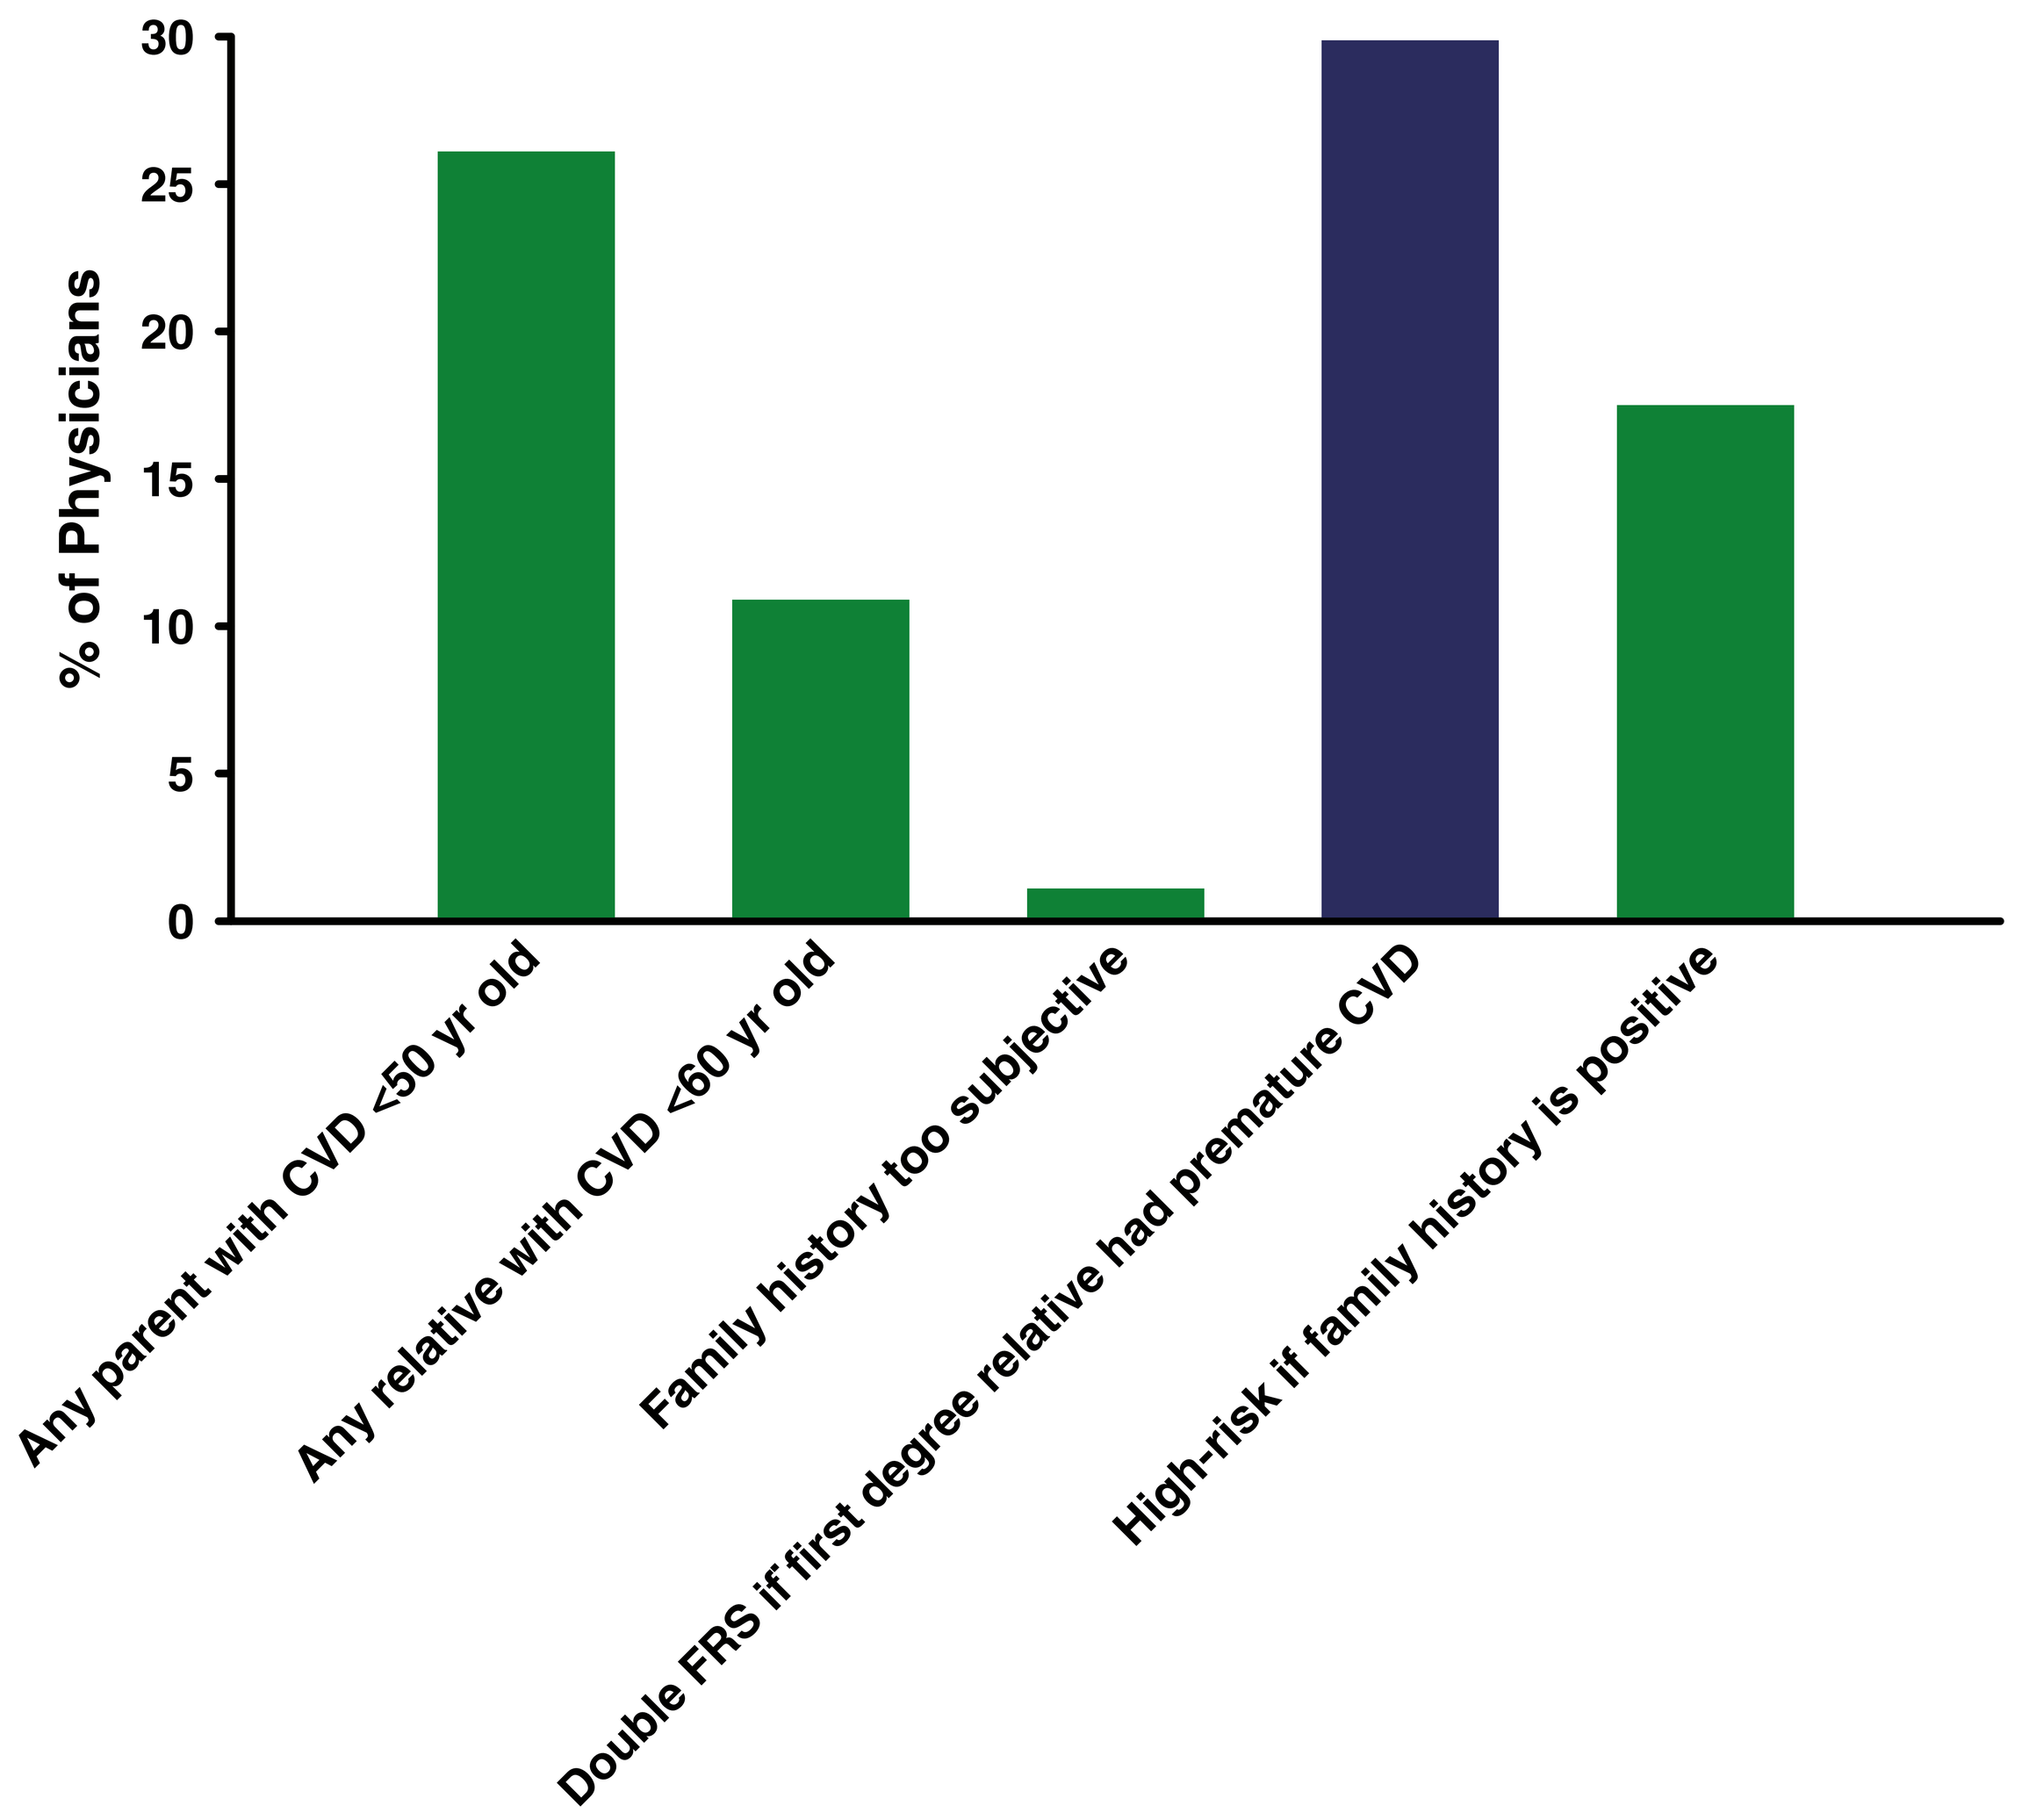
% of Physicians
0
5
10
15
20
25
30
Any parent with CVD <50 yr old
Any relative with CVD <60 yr old
Family history too subjective
Double FRS if first degree relative had premature CVD
High-risk if family history is positive

Fig. 4. Responses to "How do you incorporate family history into CV risk stratification?"

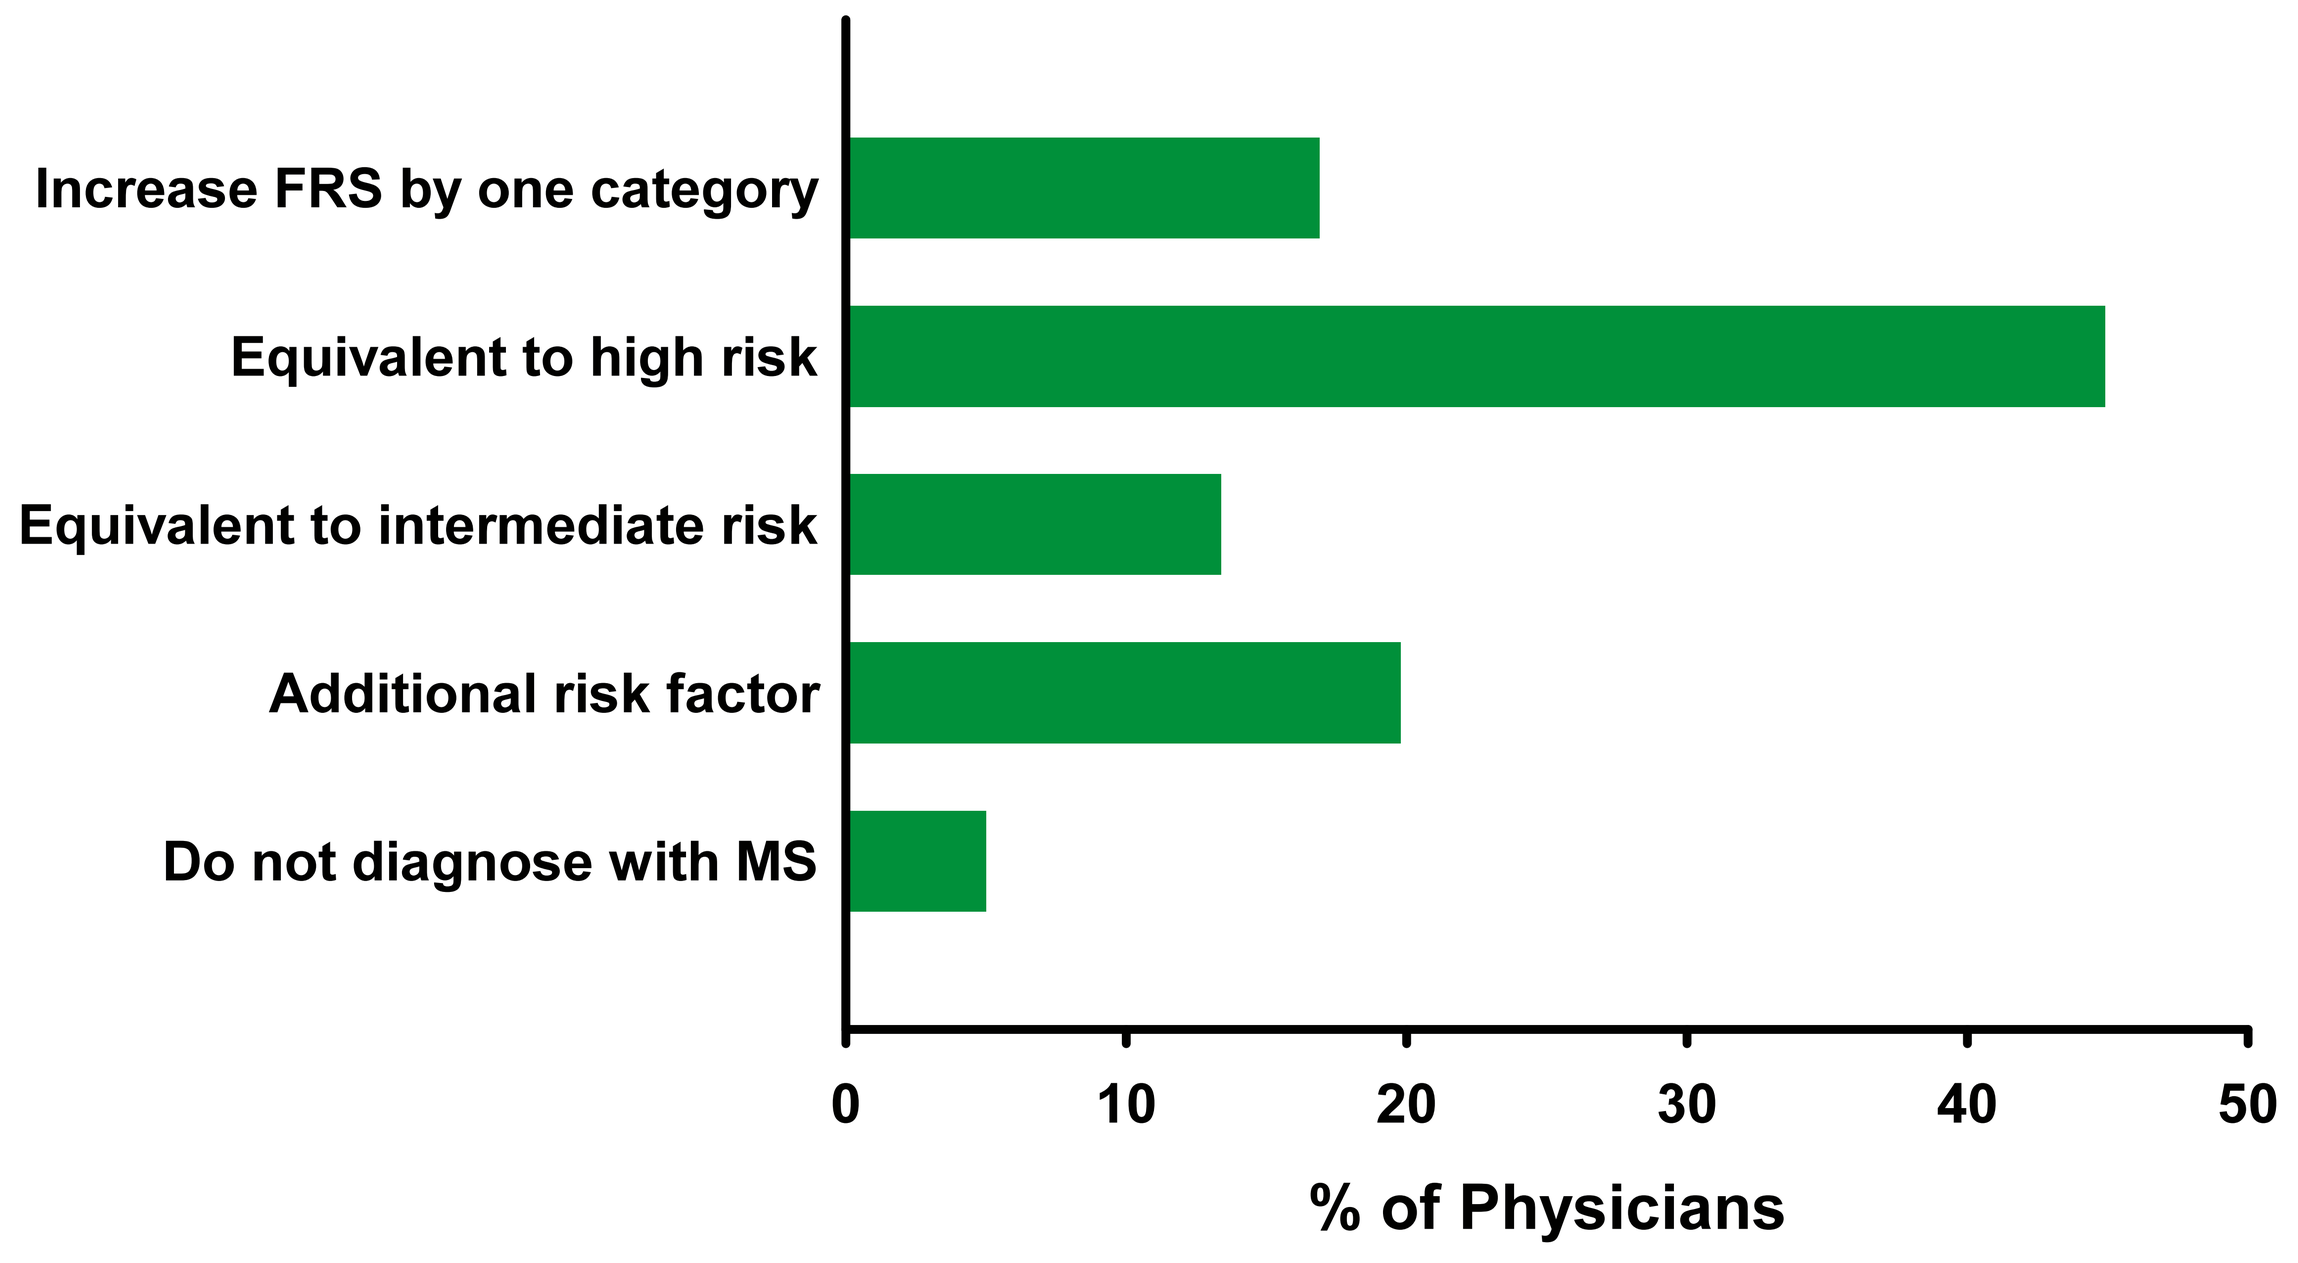
Increase FRS by one category
Equivalent to high risk
Equivalent to intermediate risk
Additional risk factor
Do not diagnose with MS
0
10
20
30
40
50
% of Physicians

Fig. 5. Responses to "How do you incorporate the metabolic syndrome (MS) in a non-diabetic patient into CV risk stratification?"

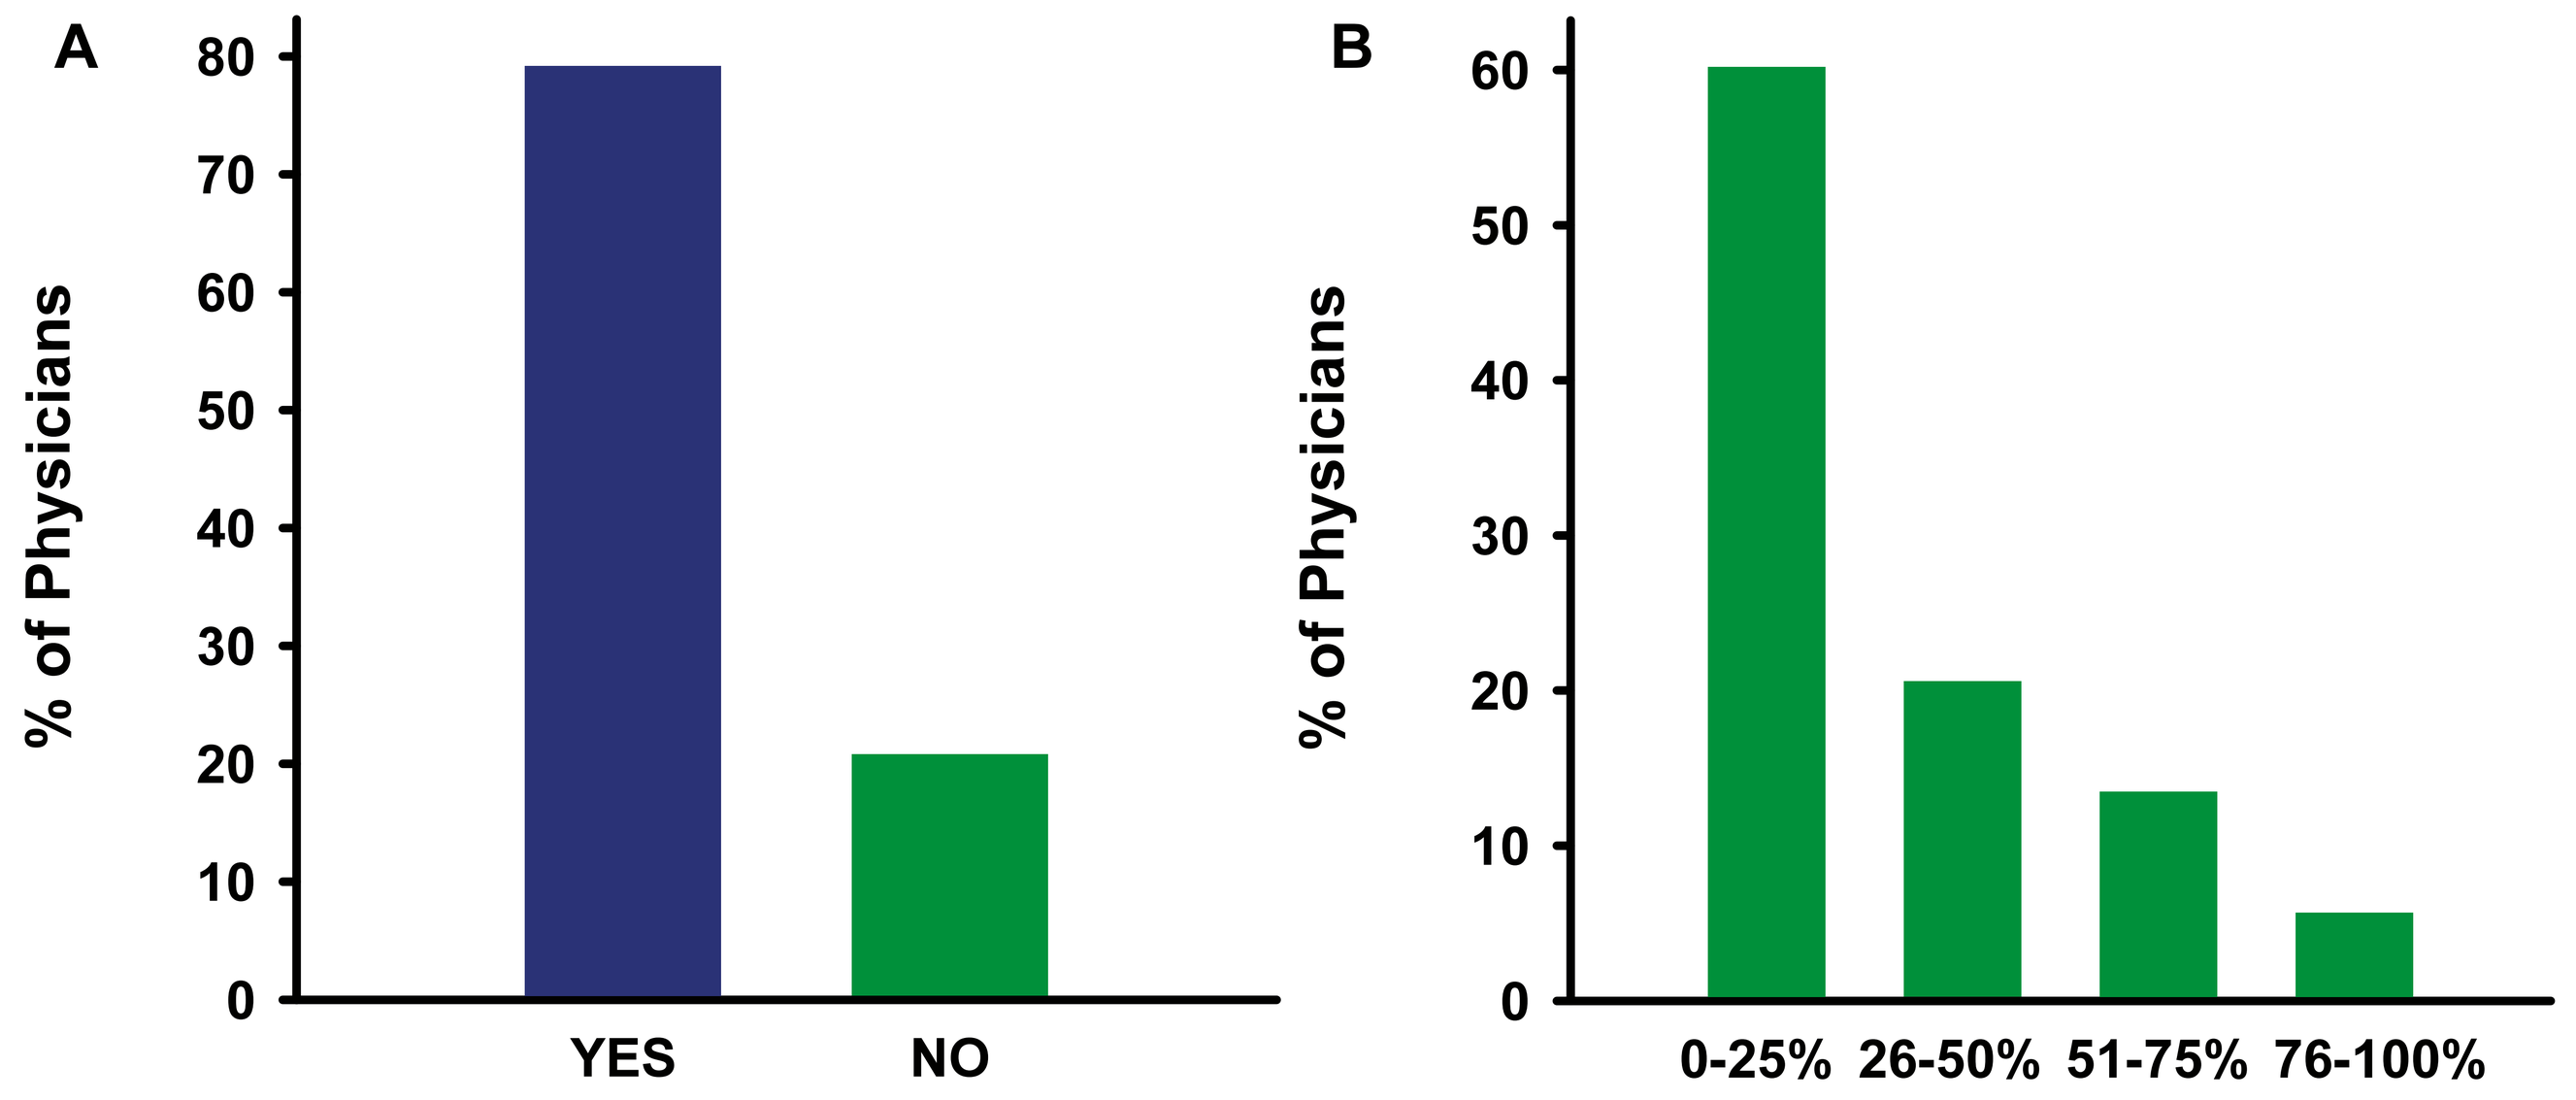
A
% of Physicians
0
10
20
30
40
50
60
70
80
YES
NO
B
% of Physicians
0
10
20
30
40
50
60
0-25%
26-50%
51-75%
76-100%

Fig. 6. Responses to (A) "Do you consider Waist Circumference to be a 'Vital Sign'?". (B) "What percentage of your patients' charts have a waist circumference documented?"

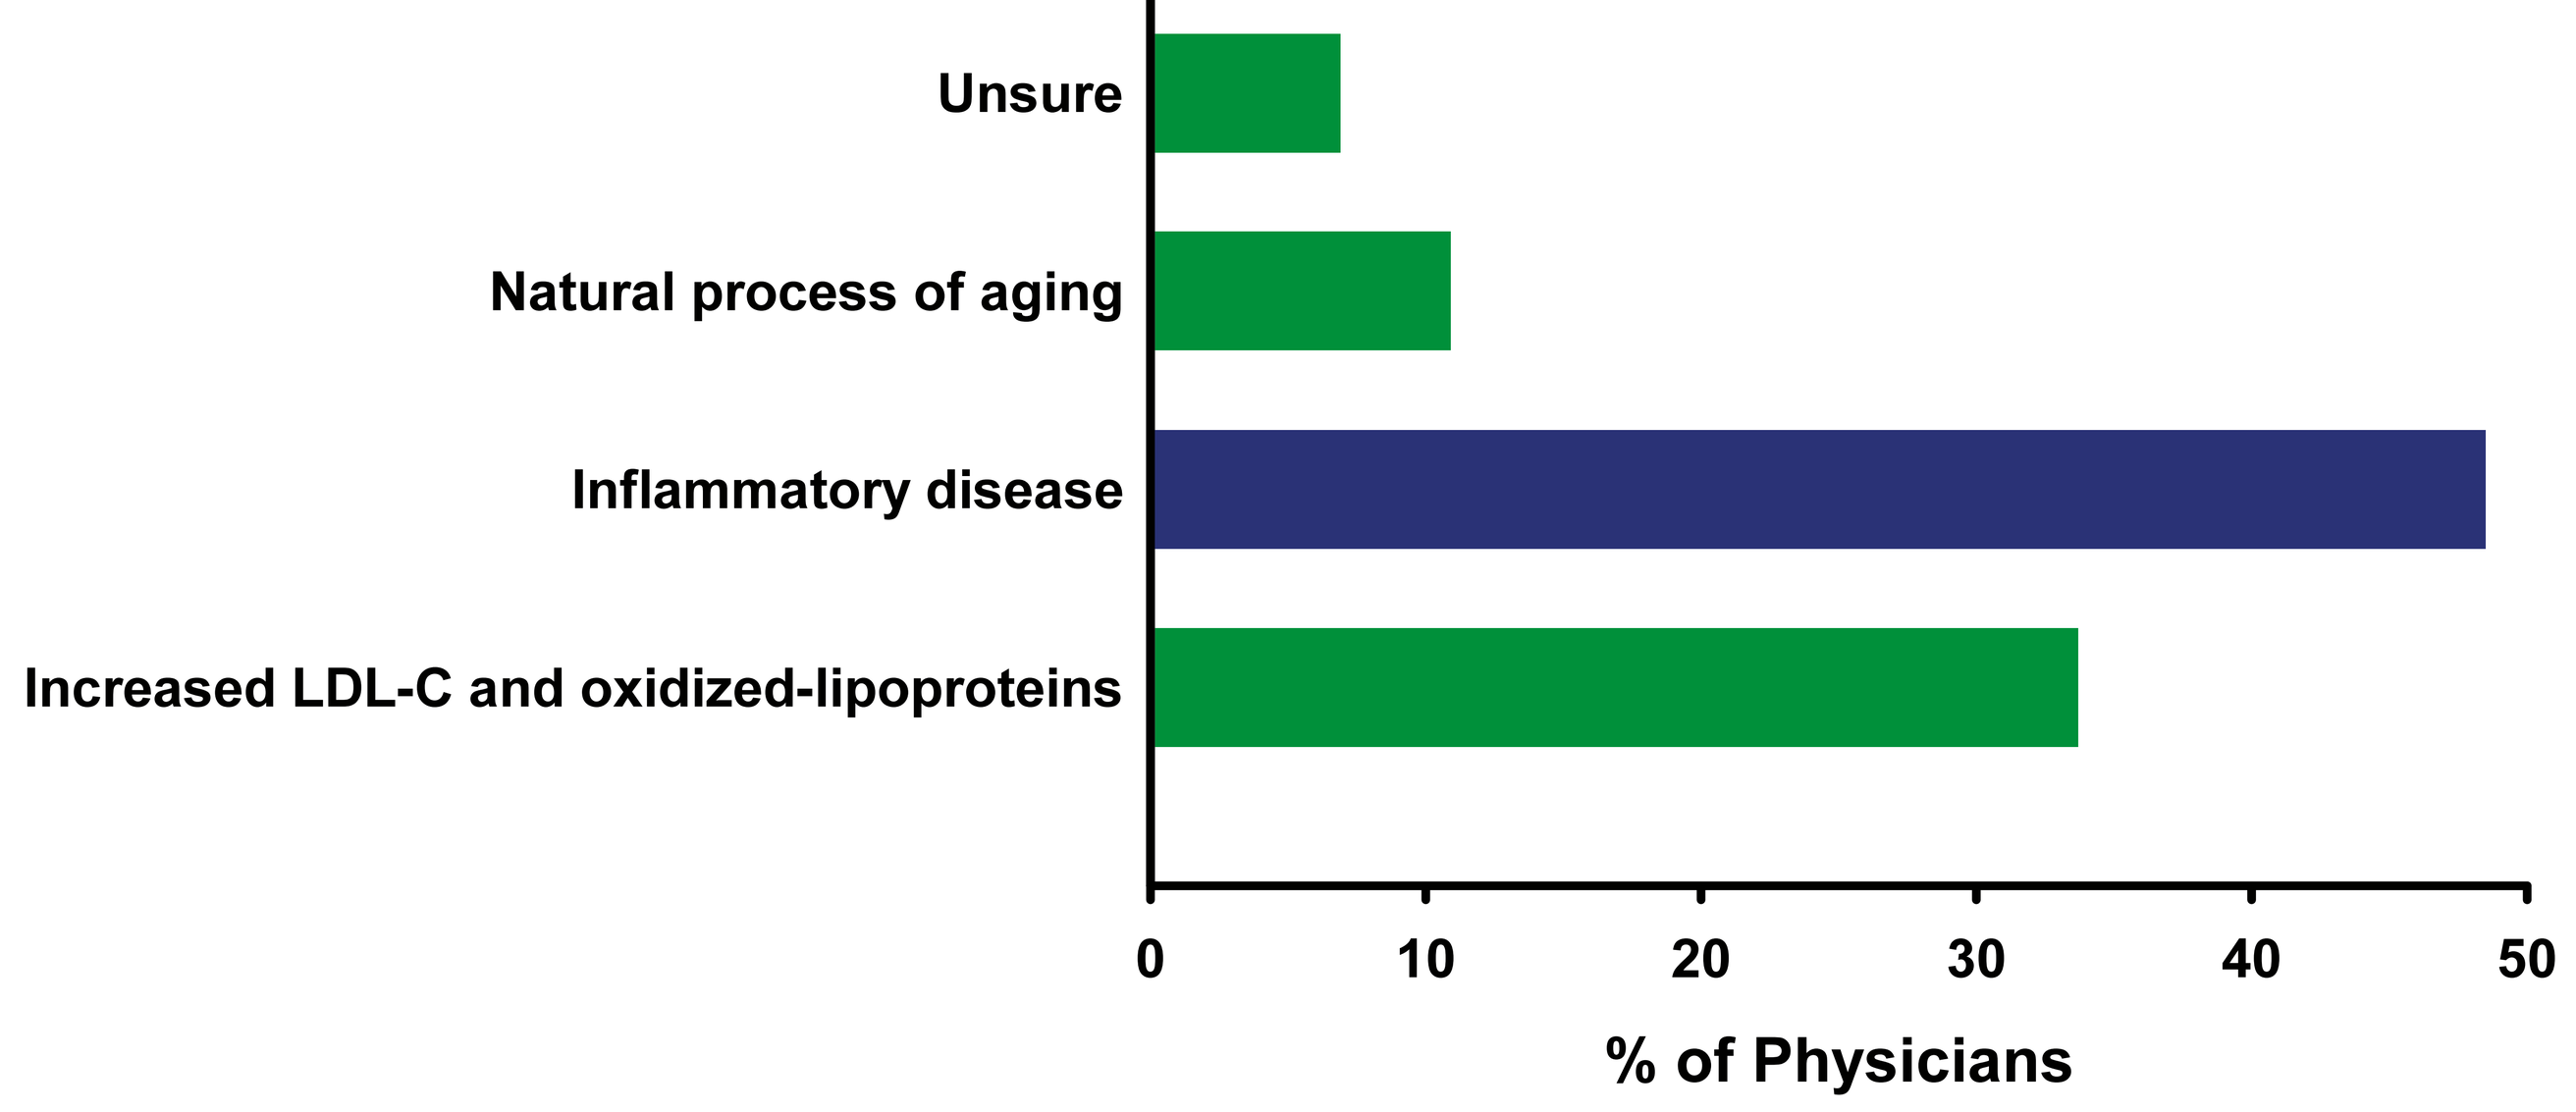
Unsure
Natural process of aging
Inflammatory disease
Increased LDL-C and oxidized-lipoproteins
0
10
20
30
40
50
% of Physicians

Fig. 7. Responses to "Which of the following statements do you think best characterizes atherosclerosis in most patients?"

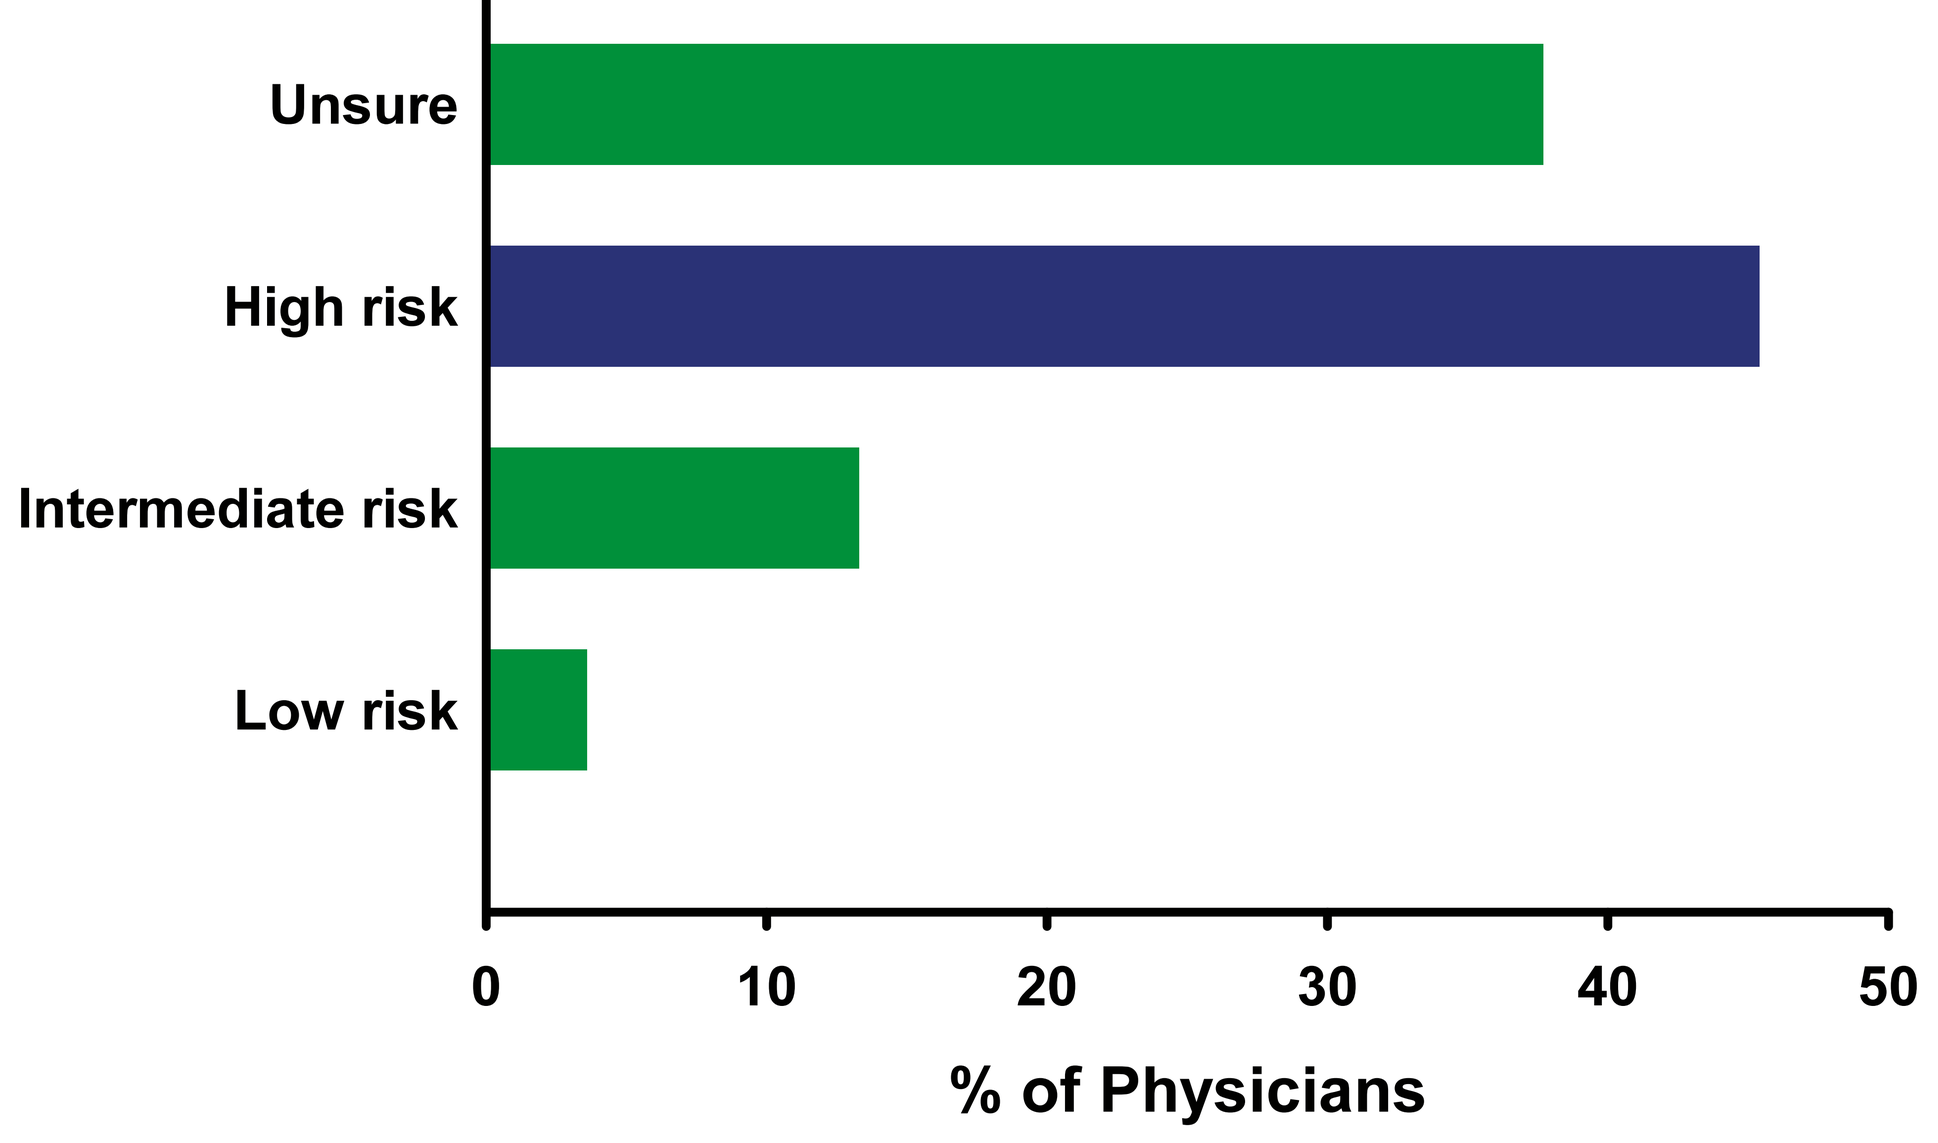
Unsure
High risk
Intermediate risk
Low risk
0
10
20
30
40
50
% of Physicians

Fig. 8. Responses to "In a 60 year old man with intermediate risk by FRS, how does a hsCRP level of 4 mg/L affect his risk stratification?"

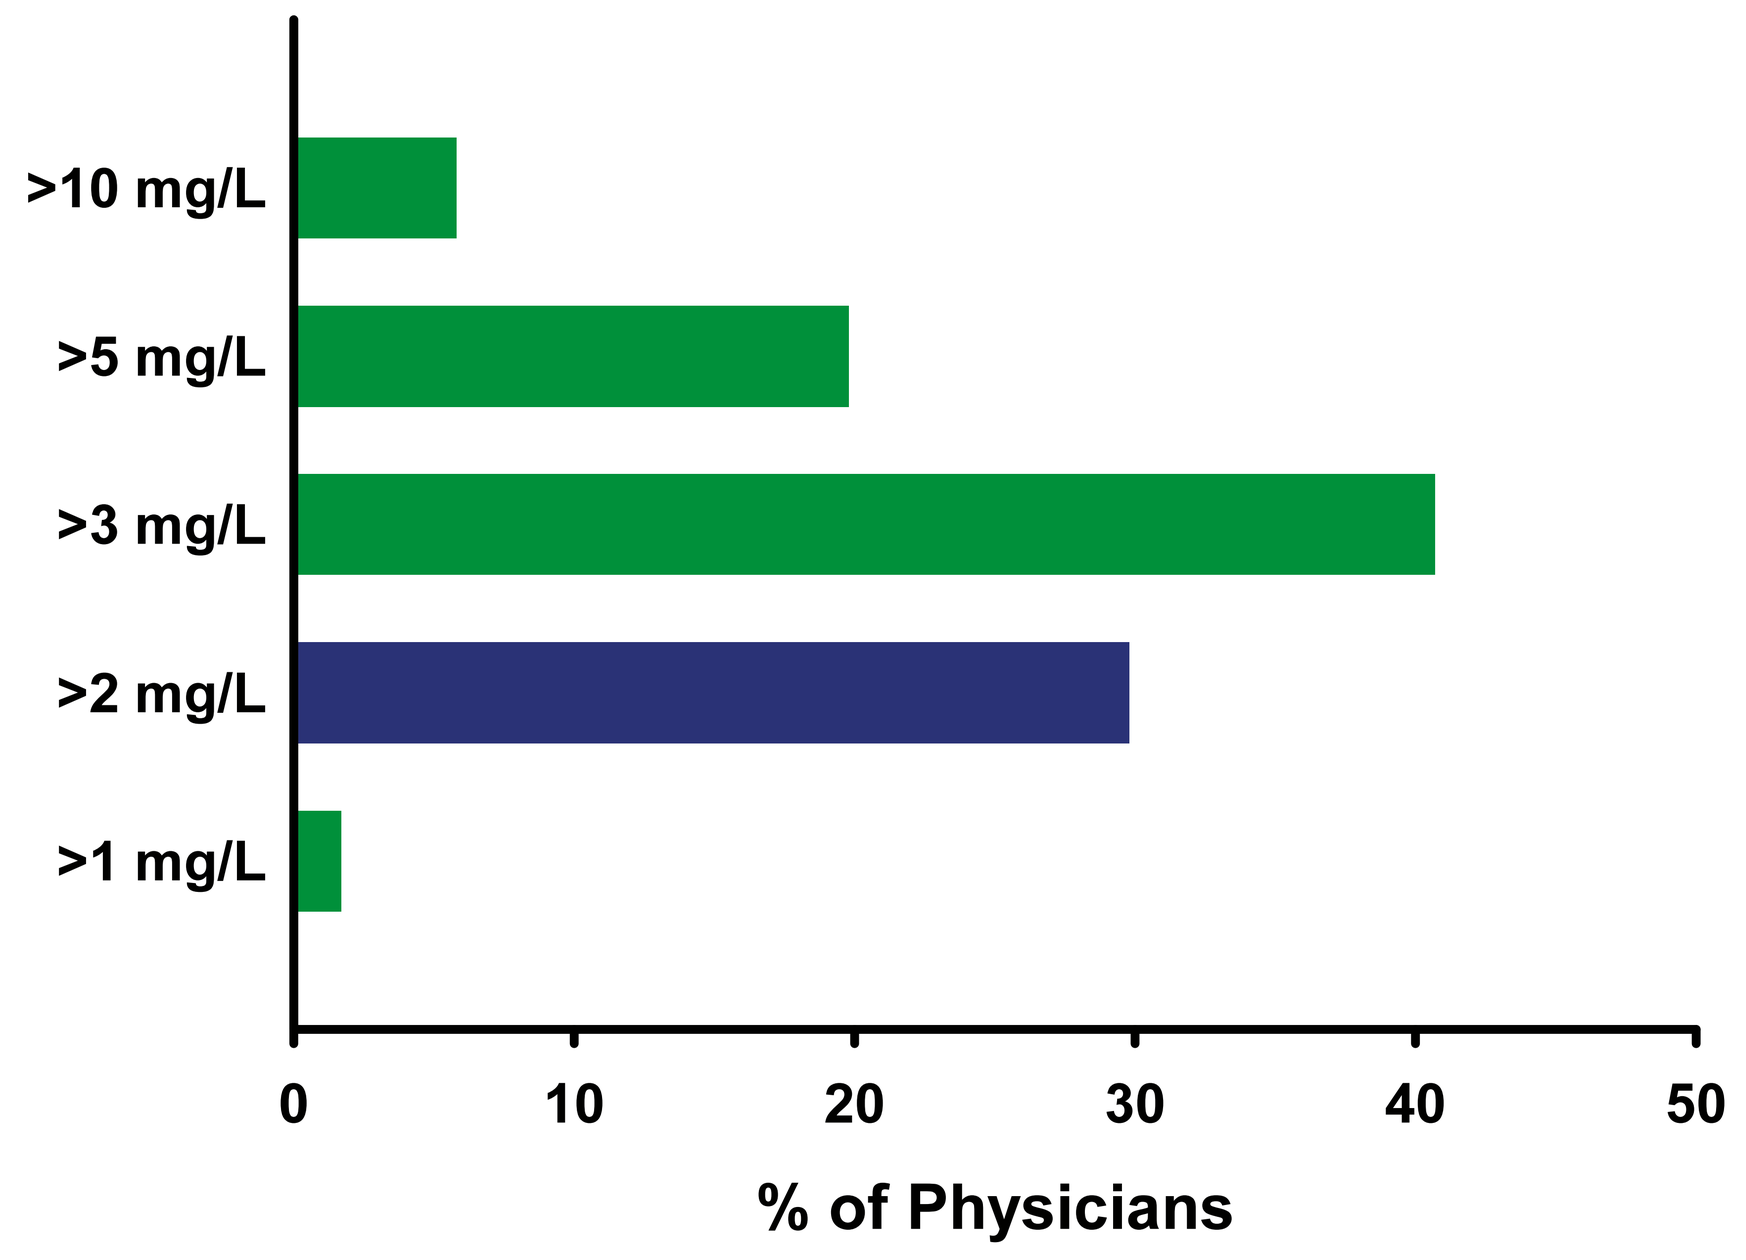
>10 mg/L
>5 mg/L
>3 mg/L
>2 mg/L
>1 mg/L
0
10
20
30
40
50
% of Physicians

Fig. 9. Responses to "You are screening an asymptomatic non-diabetic 66-year old male for future vascular risk in your office. He is a non-smoker. His LDL cholesterol is 3.2 mM (124 mg/dL) and total cholesterol 5.1 mM (197 mg/dL). His BMI is 28 and his blood pressure is 124/70 mmHg. He currently takes ASA 81 mg/dy. There is no family history of early atherosclerosis. If you would measure hsCRP, at what level of CRP would you consider statin therapy?"

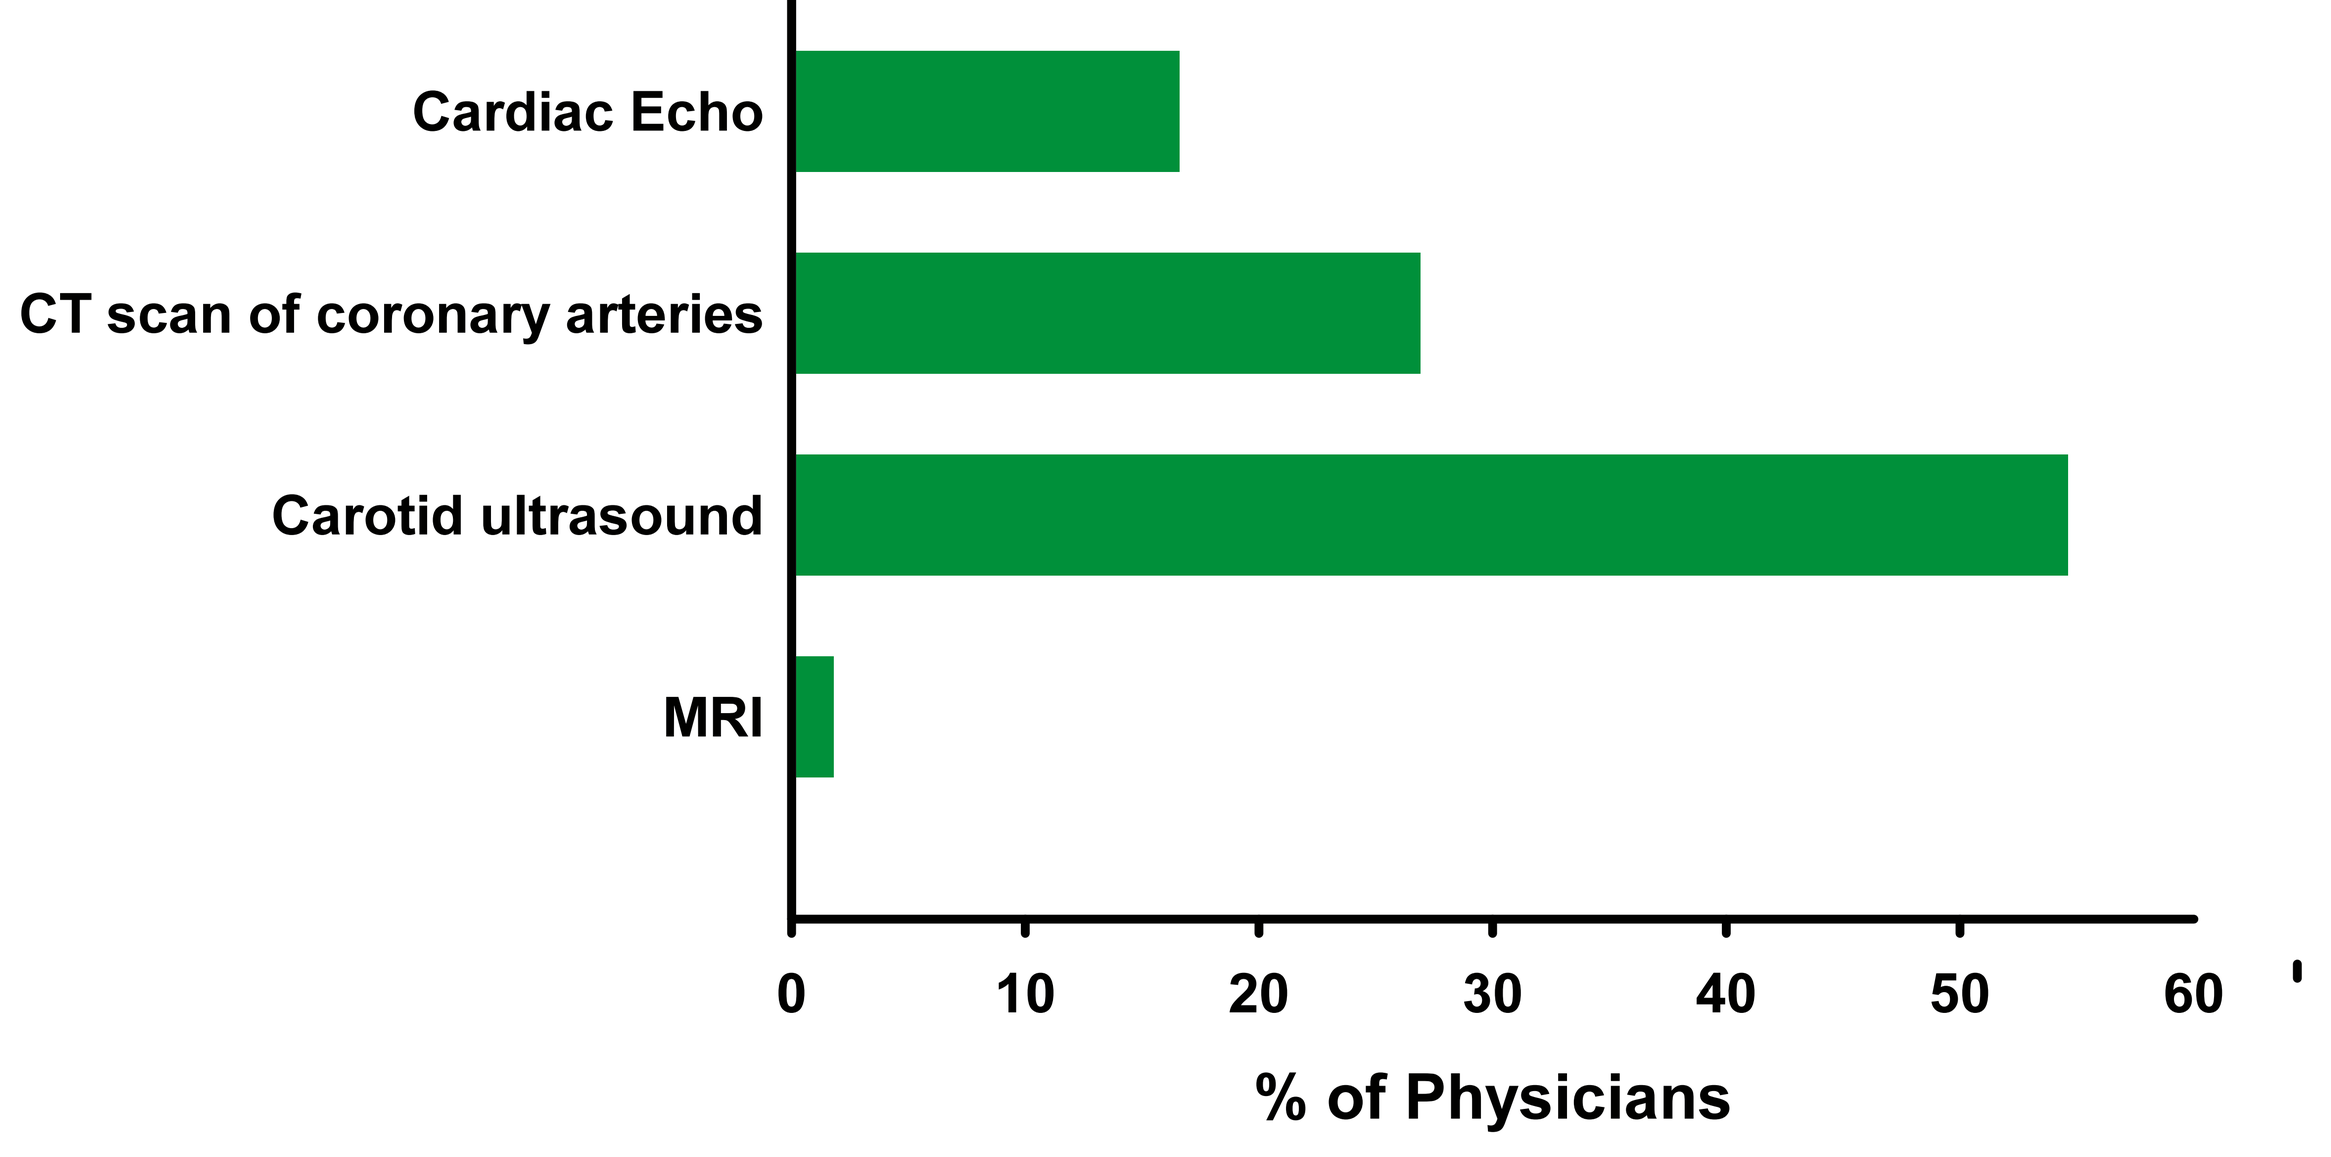
Cardiac Echo
CT scan of coronary arteries
Carotid ultrasound
MRI
0
10
20
30
40
50
60
% of Physicians

Fig. 10. Responses to "In addition to biochemical assessments, what imaging test do you believe is best suited for screening in primary prevention?"

SUMMARY

- ◆ 89% of physicians were aware of the outcomes of FRS estimates but 29.5% could not characterize FRS thresholds for high risk
- ◆ Family history was considered by 25.7% of physicians to be the most important single risk factor but only 44.4% correctly used a positive family history to roughly double the FRS, as per the Canadian Lipid Guidelines
- ◆ 79.2% of physicians considered waist circumference as a vital sign but only 5.7% routinely documented this measurement
- ◆ 44.9% and 13.4% of physicians respectively equated metabolic syndrome with high and intermediate risk
- ◆ 99.3% of physicians surveyed had heard of hs-CRP in the context of cardiovascular disease
- ◆ 54.6% of physicians surveyed were unsure how to or incorrectly re-classified a hypothetical intermediate risk patient with an elevated level of hs-CRP
- ◆ 54.6% of physicians considered carotid ultrasound the best imaging technique for screening in primary prevention

CONCLUSIONS

Despite the wide dissemination of lipid guidelines, significant knowledge gaps persist regarding optimal methods for CV risk assessment.

ACKNOWLEDGEMENTS

This work was supported by an investigator-initiated research grant from AstraZeneca Canada to M Gupta and S Verma.

www.ccrnmd.com

CCRN Canadian Cardiovascular Research Network